Miniature Check Valve Incorporated Silicone-Based Osmotic Drug Delivery System for Intravesical Drug Delivery to the Bladder

Presenting Author: Emily Abbate, TARIS Biomedical LLC, USA
Co-Authors: Heejin Lee, Sarah Hocking, Philip Jacobson, Karen Daniel, Hong Linh Ho Duc, Abizer Harianawala, TARIS Biomedical LLC, USA

Objectives

1. Identify factors affecting the performance of miniature check valve.
2. Explain operating principles of a tube-shaped implantable osmotic pump.
3. Determine design considerations for long-term intravesical drug delivery system.
4. Identify dominant factors affecting the drug release of a silicone-based osmotic pump integrated with miniature one-way valves.

Background

Sub-millimeter scale orifices, channels, and/or micro-valves are often used as drug release passages in reservoir-based implantable osmotic drug delivery systems. The drug delivery systems function is dependent on both outlet patency and robustness against clogging and damage. Additionally, key attributes for scalable manufacturing of the system is the cost-effectiveness and ease of integration of these outlets. Based on these considerations, we present an implantable osmotically driven silicone based drug delivery system with miniature check valve(s) to achieve continuous local drug delivery of Molecule X to the bladder.

Drug Delivery System

Mini-tablets containing Molecule X were loaded into a lumen of a dual-lumen silicone tube that is semi-permeable to water and impermeable to drug. Then a bi-oval shaped superelastic nitinol wireform was inserted into the second lumen to impart a retentive shape. One or more parylene-coated cylindrical silicone components were placed and integrated within the lumen containing Molecule X (drug reservoir). The number and length of these drug release outlets were readily adjustable. Prototype systems with multiple design options were built and terminally sterilized through gamma irradiation. Then, *in vitro* characterization studies were performed past 30-days to evaluate the osmotically driven drug release. The number and length of the drug release outlets was tested along with two different tubing thicknesses. Collected timepoint samples were analyzed using HPLC.

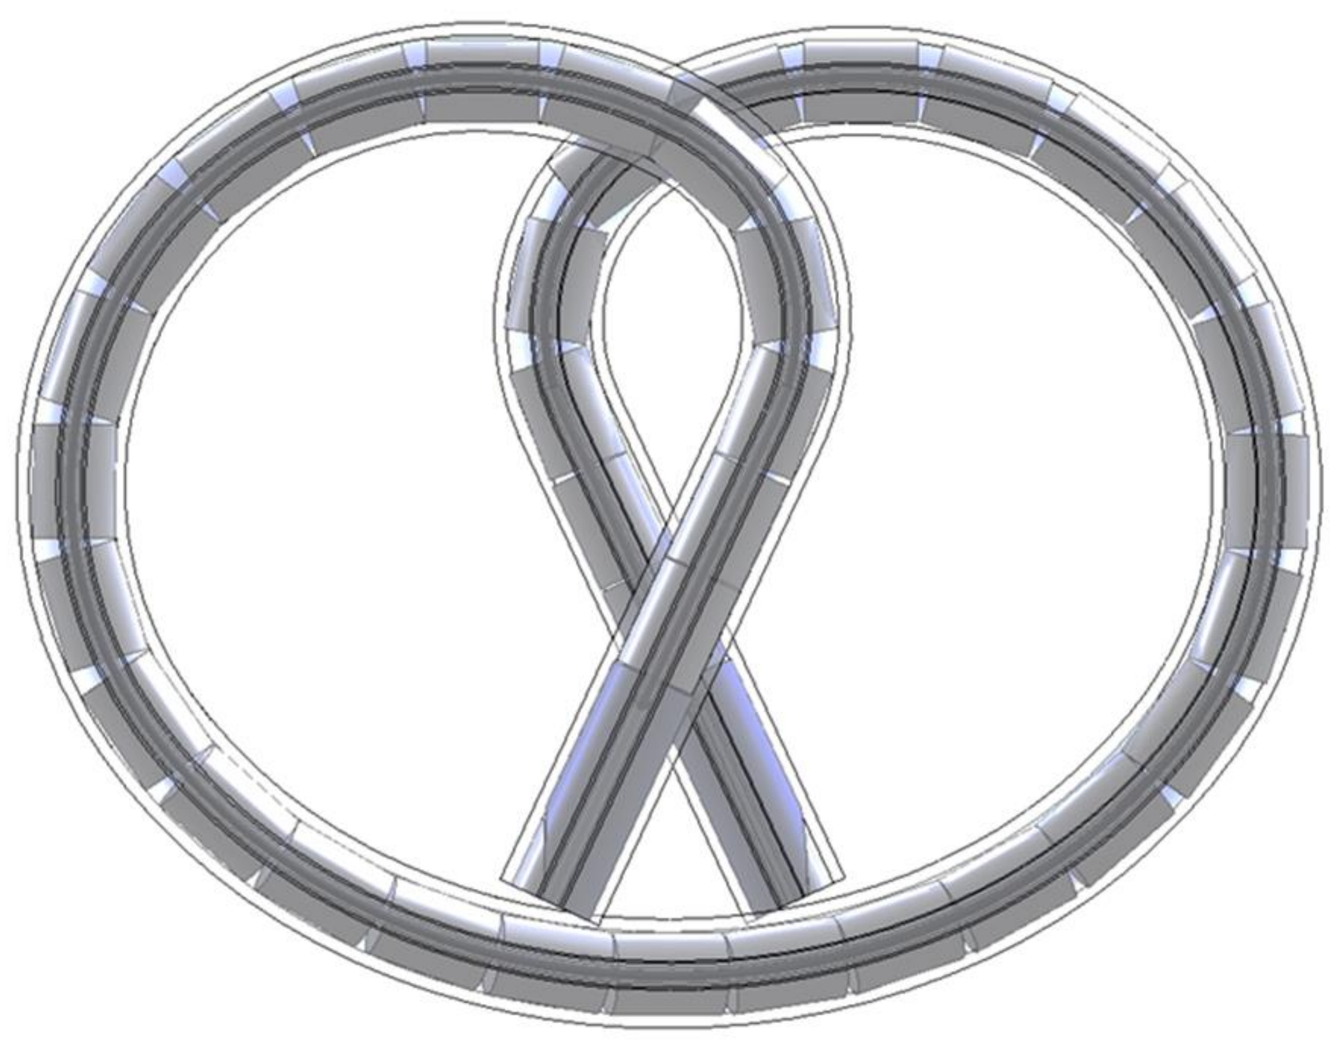

Figure 1: Implantable Silicone based Drug Delivery System

Miniature Check Valve Mechanism

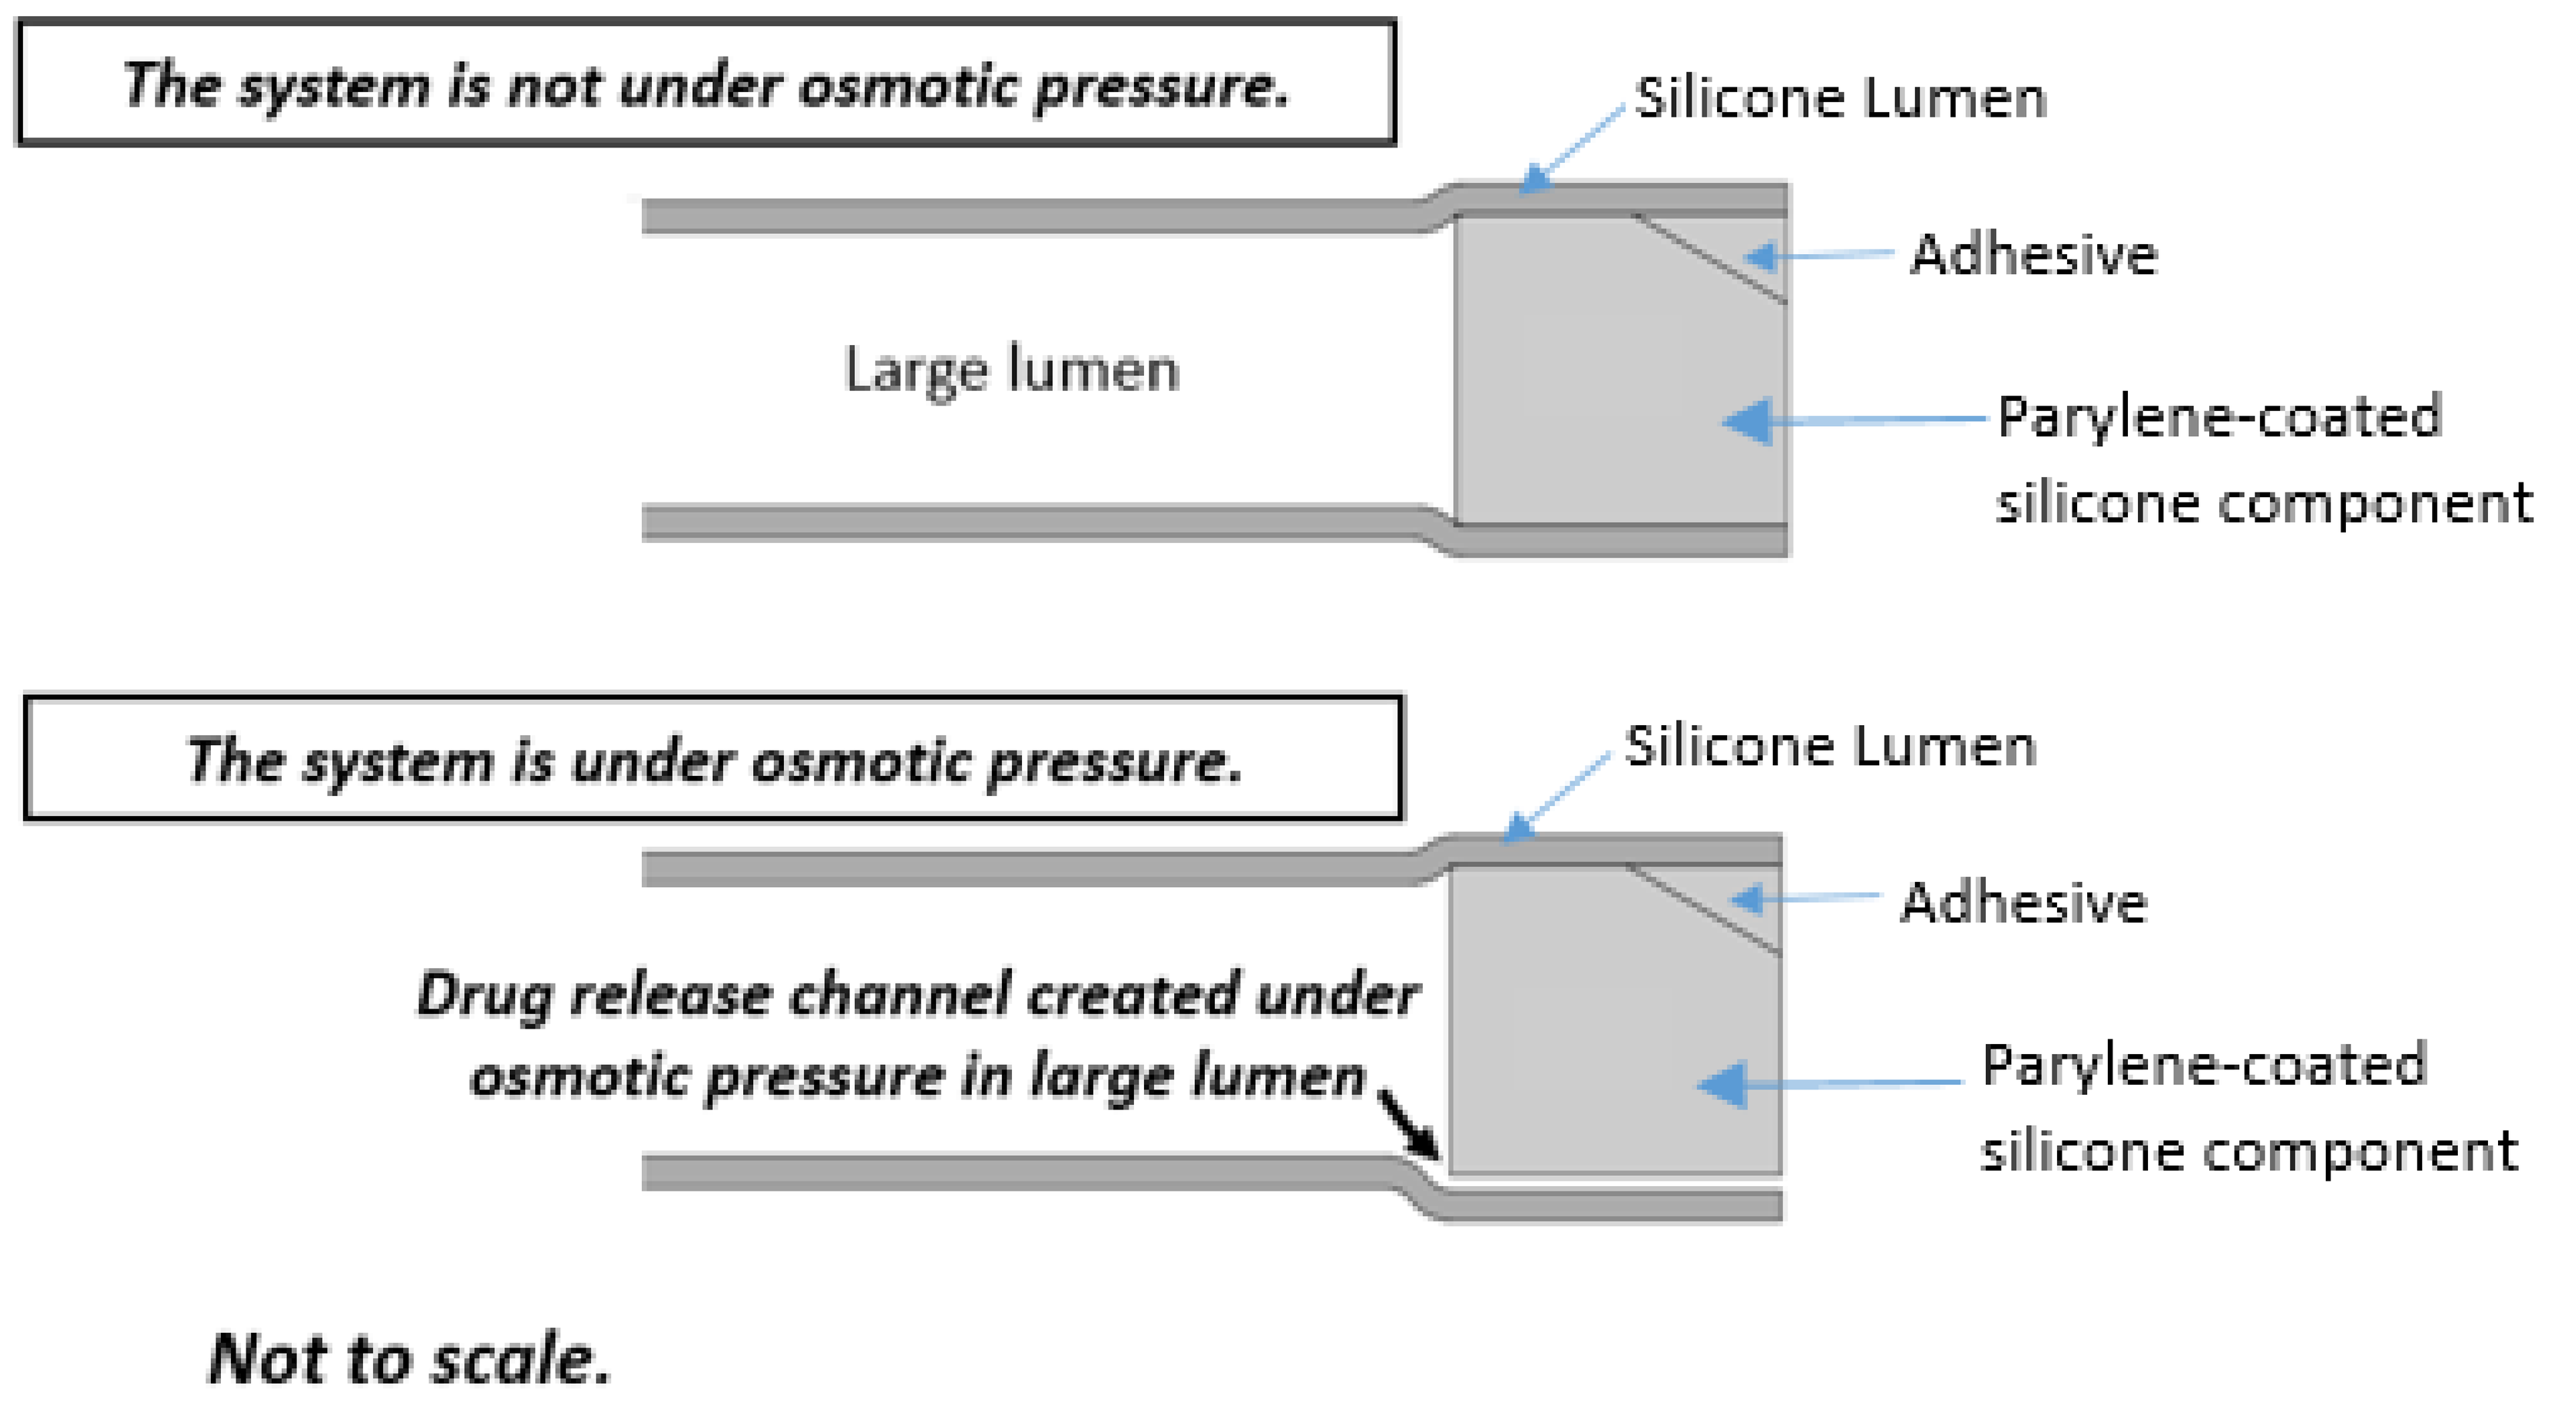
The system is not under osmotic pressure.
Silicone Lumen
Adhesive
Large lumen
Parylene-coated silicone component
The system is under osmotic pressure.
Silicone Lumen
Adhesive
Drug release channel created under osmotic pressure in large lumen
Parylene-coated silicone component
Not to scale.

Figure 2: Miniature Check Valve Incorporated Silicone Based-Osmotic Drug Delivery System Before and During Drug Release

In Vitro Characterization

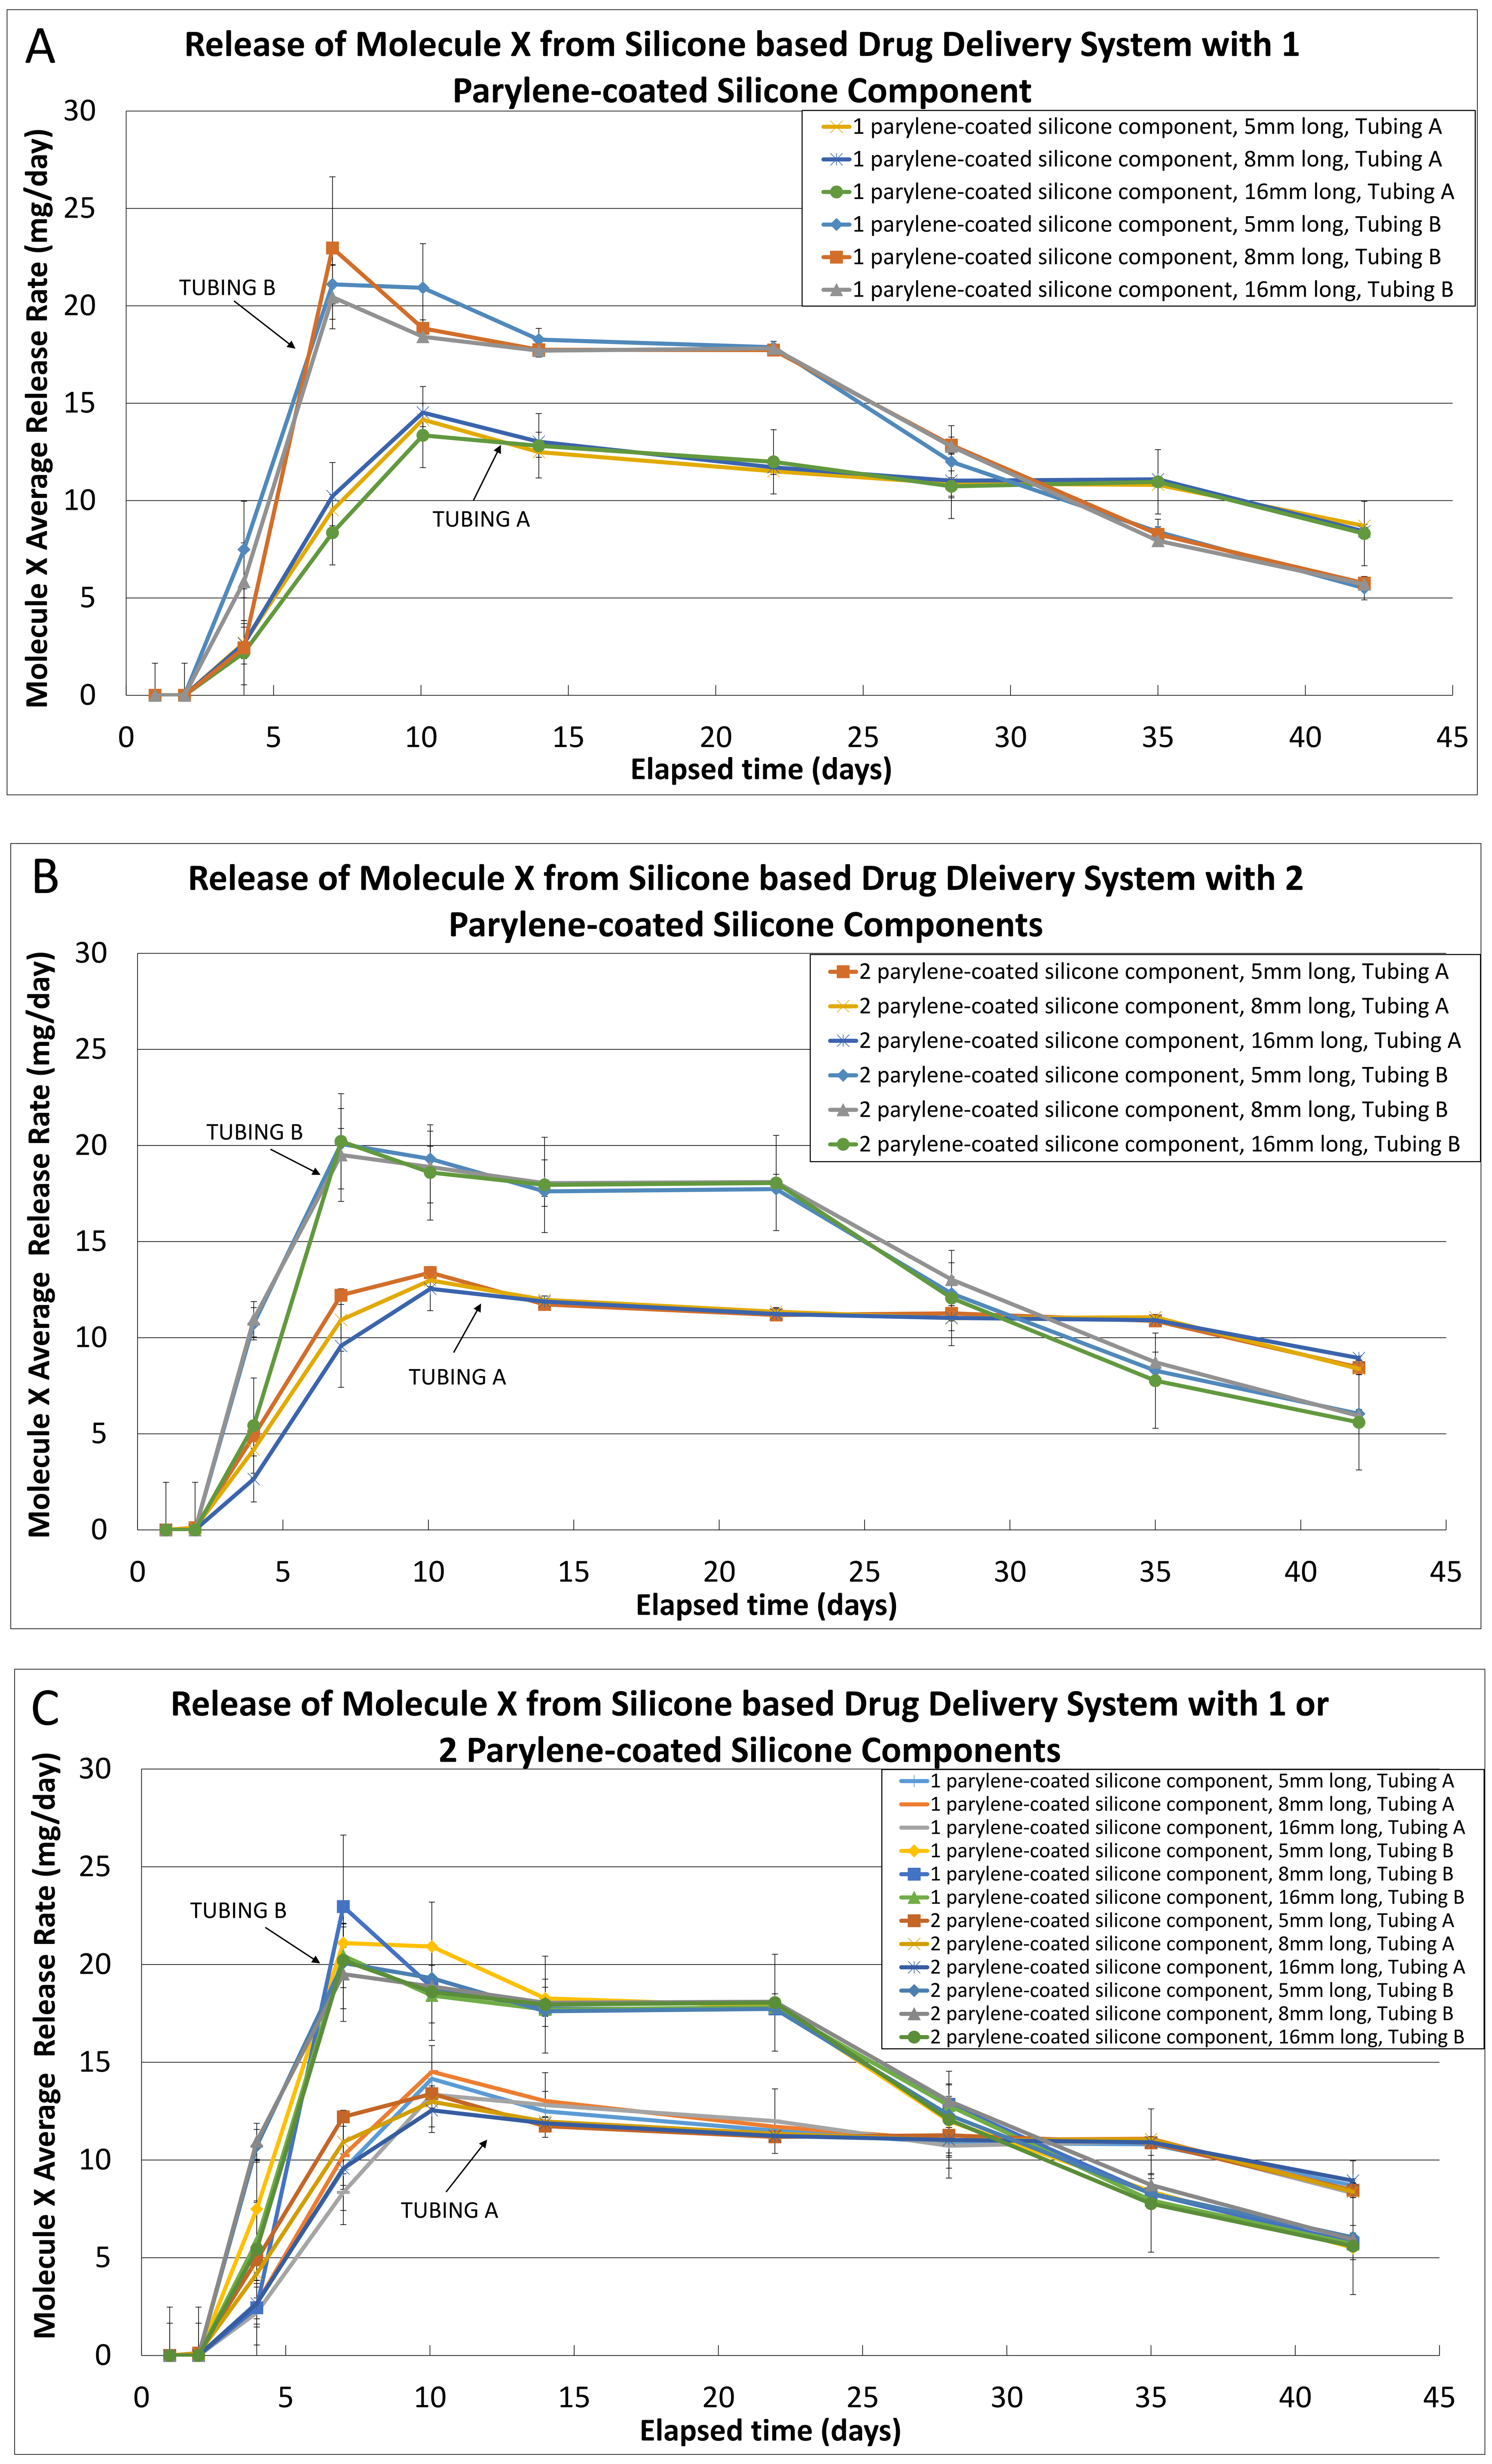
A
Release of Molecule X from Silicone based Drug Delivery System with 1 Parylene-coated Silicone Component
Molecule X Average Release Rate (mg/day)
Elapsed time (days)
TUBING B
TUBING A
1 parylene-coated silicone component, 5mm long, Tubing A
1 parylene-coated silicone component, 8mm long, Tubing A
1 parylene-coated silicone component, 16mm long, Tubing A
1 parylene-coated silicone component, 5mm long, Tubing B
1 parylene-coated silicone component, 8mm long, Tubing B
1 parylene-coated silicone component, 16mm long, Tubing B
B
Release of Molecule X from Silicone based Drug Dleivery System with 2 Parylene-coated Silicone Components
Molecule X Average Release Rate (mg/day)
Elapsed time (days)
TUBING B
TUBING A
2 parylene-coated silicone component, 5mm long, Tubing A
2 parylene-coated silicone component, 8mm long, Tubing A
2 parylene-coated silicone component, 16mm long, Tubing A
2 parylene-coated silicone component, 5mm long, Tubing B
2 parylene-coated silicone component, 8mm long, Tubing B
2 parylene-coated silicone component, 16mm long, Tubing B
C
Release of Molecule X from Silicone based Drug Delivery System with 1 or 2 Parylene-coated Silicone Components
Molecule X Average Release Rate (mg/day)
Elapsed time (days)
TUBING B
TUBING A

Figure 3: Average Release Rate (mg/day) of Molecule X in Tubing A or B (A) with 1 parylene-coated silicone component, (B) with 2 parylene-coated silicone components, and (C) Overlap of (A) and (B)
n=3 per sample type

Results and Conclusions

Simple miniature check valves were designed that could be integrated into an implantable silicone-based tubular osmotic system for intravesical drug delivery. This design produced a new zero-order drug release profile that extends beyond 30 days with Molecule X, which was formulated to have sufficient osmotic pressure to reach cracking pressure of the embedded check valve design. It was determined that the number and length of the check vales had no impact on the drug release profile, but that an alteration to tubular wall thickness could decrease or increase peak release. Overall, this design has potential employment in other implantable drug delivery systems, and the design is simple and scalable.

Acknowledgements

We would like to thank everyone from the TARIS Biomedical LLC team for their efforts and contributions to this project.